A PHASE I, OPEN-LABEL, 2-PART STUDY TO ESTABLISH ABSOLUTE BIOAVAILABILITY AND THE ADME OF EVOGLIPTIN IN HEALTHY MALE SUBJECTS BY A LIGHT-LABEL APPROACH.

I. Shaw[2], Dae Young Lee[1], Hyunjoo Shim[1], Ji Su Song[1], H Walker[2], J Collier[2], Sookie La[3], Hee Joo Lee[3], Stephen R. Dueker[3]

Dong-A ST Co.,Ltd[1] (Seoul, Republic of Korea), Quotient Clinical Ltd[2] (Nottingham, United Kingdom), BioCore[3] (Seoul, Republic of Korea)

INTRODUCTION

Evogliptin is a new treatment for Diabetes mellitus type 2 (T2DM). It inhibits the serine protease dipeptidyl peptidase 4 (DPP4), which is in charge of degradation of glucagon like peptide 1 (GLP1), a 30-amino acid peptide hormone secreted by intestinal L-cells in response to meal ingestion, which stimulates insulin secretion from β-cells while inhibiting hepatic glucose production. Active GLP1 is rapidly converted to inactive GLP1 by DPP4, thus limiting its therapeutic practicality. Therefore, inhibition of DPP4 can increase the levels of endogenous intact GLP1.

Intravenous (IV) microtracer dosing is an established tool for absolute bioavailability assessment. It involves the concomitant administration of an IV microdose of a drug, labelled with microtracer amounts (not more than (NMT) 37KBq (1000 nCi) of ^{14}C, with an oral therapeutic dose of the drug. The IV dose is administered via a short infusion, which terminates at the Tmax of the oral drug. Using this approach the preclinical IV toxicology requirements are reduced and the IV formulation is simple compared to that required for a therapeutic IV dose. Oral tracer dosing, which utilisers an oral therapeutic dose of parent drug, labelled with microtracer amounts of ^{14}C, has also been utilised successfully to evaluate absorption, metabolism, distribution and elimination (ADME) investigations. Utilising mictrotracer amounts of radioactivity means that it is not necessary to have a formal dosimetry assessment prior to dosing, and as such the time it takes to establish and conduct an ADME programme can be substantially reduced, compared to a standard human ADME study.

This poster describes a 2 part study combing an IV microtracer study (part 1) with an oral tracer limb (part 2) to successfully measure the absolute bioavailability and the mass balance of [^{14}C]-Evogliptin, and investigate the metabolism of the drug.

METHODS

This was a single centre, open-label, non-randomised, 2-part study in healthy male subjects. Twelve subjects were enrolled in the study; 6 subjects in Part 1 and 6 subjects in Part 2. Subjects were screened for eligibility to participate in the study within 28 days before dosing. Approvals were obtained from Ethics, Medicines and Healthcare products Regulatory Agency prior to subject recruitment. The study outline is described in Figure 1.

The primary objectives were:

- To determine the absolute bioavailability of Evogliptin in healthy subjects
- To assess the absorption, metabolism and excretion of Evogliptin after oral administration of carbon-14 labelled [^{14}C]-Evogliptin to healthy subjects

Figure 1

Part 1 Schedule	Part 2 Schedule
Screening **Day -28 to Day -2**	**Screening** **Day -28 to Day -2**
↓	↓
Admission: Day -1	**Admission: Day -1**
↓	↓
Day 1 **Regimen A:** a single oral therapeutic dose of a 5 mg Evogliptin tablet followed by **Regimen B:** an IV microdose of 20 µg [^{14}C]-Evogliptin containing NMT 37.0 kBq [1000 nCi] ^{14}C administered as a 10 min infusion ending at 4 h post-oral dose	**Day 1** **Regimen C:** a single oral therapeutic dose of 5 mg [^{14}C]-DA-1229 containing NMT 37.0 kBq [1000 nCi] ^{14}C, administered as a solution
↓ **Discharge: Day 3** ↓	↓
Return Visits **Days 4, 6, 8 and 11**	**Discharge Day 11**

Samples for the analysis of IV and oral PK in plasma, total radioactivity (TRA) in whole blood and plasma, metabolite profiling and identification in plasma, and for haematology and clinical chemistry safety assessments, were taken at specified time points. Urine and faecal samples for analysis of TRA were also taken at specified time points.

- The clinical study including development and manufacture of the IV and oral ^{14}C drug products was performed at Quotient Clinical.
- Sample analysis by LC MS/MS for Evogliptin and known metabolites was performed by BioCore.
- Analysis by AMS for TRA (parts 1 & 2), and ^{14}C-Evogliptin (part 1) in plasma and TRA in whole blood, urine and faeces (part 2) was performed by Eckert & Ziegler Vitalea Science (EZVS).
- Metabolite profiling was performed by EZVS with Metabolite identification at BioCore.

After processing for analysis, samples for AMS analysis were graphitised according to standard laboratory procedures and then analysed for ^{14}C contents by AMS. Sample analysis criteria and specifications were identified in laboratory standard operating procedures. Accuracy acceptance criteria for the absolute bioavailability schedule was set at +/-15% of nominal values for standards and QC samples except at the LLOQ which was set at +/-20%.

Total Carbon for urine and faeces was determined via isotope dilution with ^{13}C-glycine and monitoring of the $^{13}C/^{12}C$ ratio simultaneously with the $^{14}C/^{13}C$ ratio.

The raw data from the AMS (^{14}C counts and ^{12}C current) for each sample was then converted through a series of qualified calculations to the final output of a value for ng-eq/ml.

Bioanalysis methods used in the establishment of absolute bioavailability were fully validated with 4 levels of QCs for batch acceptance criteria.

RESULTS AND DISCUSSION

Evogliptin was well tolerated. There were no deaths, severe AEs or serious AEs reported during the study, and no subject was withdrawn as a result of an AE. There were no clinically significant findings in any laboratory assessments, vital signs, ECGs or physical examinations.

Part 1

The geometric mean (CV%) of key PK parameters of Evogliptin, [^{14}C]-Evogliptin and TRA following a 5 mg oral dose of DA-1229 co-administered with a single IV Infusion of 20 µg [^{14}C]-DA-1229 over 10 min, are presented in Table 1. Following oral administration Evogliptin absorption was prolonged; median Tmax was 5h. Absolute bioavailability of Evogliptin was 50.2%. The ratio of AUC(0-inf) [14C]-DA-1229:TRA was approximately 77 % which suggests that there is metabolism and/or formation of breakdown products following IV administration of [14C]-DA-1229.

Table 1

Regimen Route Dose Analyte No. of Subjects	A Oral 5 mg Evogliptin N = 6	B IV 20 µg [^{14}C]-Evogliptin N = 6	B IV 20 µg Total Radioactivity N = 6
Tmax (h) (range)	5.00 (2.00-10.00)	0.17 (0.08-0.17)	0.17 (0.08-0.17)
Cmax (ng/mL)[a]	4.57 (20.4)	0.781 (28.1)	0.756 (17.7)
AUC(0-last) (ng.h/mL)[a]	189 (23.5)	1.51 (30.8)	1.91 (28.5)
AUC(0-inf) (ng.h/mL)[a]	194 (24.3)	1.56 (31.3)	2.02 (27.7)
T1/2 (h)	53.59 (15.3)	46.53 (27.4)	45.19 (27.5)
MRT (h)	55.09 (15.9)	41.61 (25.2)	39.76 (27.1)
CL (mL/min)	NC	216 (31.4)	NC
Vz (L)	NC	868 (10.1)	NC
Vss (L)	NC	538 (16.2)	NC
F(0-last) (%)	50.220 (17.3)	NC	NC
F(0-inf) (%)	50.247 (17.1)	NC	NC

[a] ng equivalent for TRA; NC: not calculated

Part 2

The geometric mean (CV%) of key PK parameters of DA-1229, M7, M8 and TRA following a single 5 mg oral dose of [^{14}C]-Evogliptin are presented in Table 2.

Table 2

Regimen Route Dose Analyte No. of Subjects	C Oral 5 mg Evogliptin N = 6	C Oral 5 mg M7 N = 6	C Oral 5 mg M8 N = 6	C Oral 5 mg Total Radioactivity N = 6
Tmax (h) (range)	6.00 (3.00-6.02)	6.01 (3.00-8.00)	6.01 (3.00-8.00)	6.00 (2.00-8.00)
Cmax (ng/mL)[a]	4.54 (14.5)	0.419 (26.8)	0.408 (33.5)	10.1 (18.9)
AUC(0-last) (ng.h/mL)[a]	196 (15.3)	9.75 (28.2)	12.1 (34.9)	370 (22.7)
AUC(0-inf) (ng.h/mL)[a]	202 (16.5)	10.3 (28.2)	12.9 (33.4)	392 (23.6)
T1/2 (h)	55.24 (8.4)	18.81 (24.6)	25.16 (32.3)	46.20 (21.8)
MRT (h)	55.08 (18.3)	25.97 (22.7)	34.79 (21.4)	53.20 (20.8)
AUC(0-inf) M/P ratio	NC	0.049 (14.5)	0.061 (17.9)	NC
Cmax M/P ratio	NC	0.089 (13.7)	0.086 (21.1)	NC

[a] ng equivalent for TRA; NC: not calculated

The mean cumulative recovery in urine and faeces is presented by time point in Table 3.

Table 3

Collection Period (h)	Mean Cumulative %Ae in Urine (%)	Mean Cumulative %Ae in Faeces (%)
-24 to 0	NC	NC
0–6	7.96	N/A
0–12	17.9	N/A
0–24	27.6	4.68
0–48	36.3	20.7
0–72	40.2	32.2
0–96	42.3	35.9
0-120	43.6	36.7
0-144	44.4	37.2
0-168	45.0	38.8
0-192	45.5	41.2
0-216	45.8	41.5
0-240	46.1	42.8
Total Recovery	88.9	

Figure 2

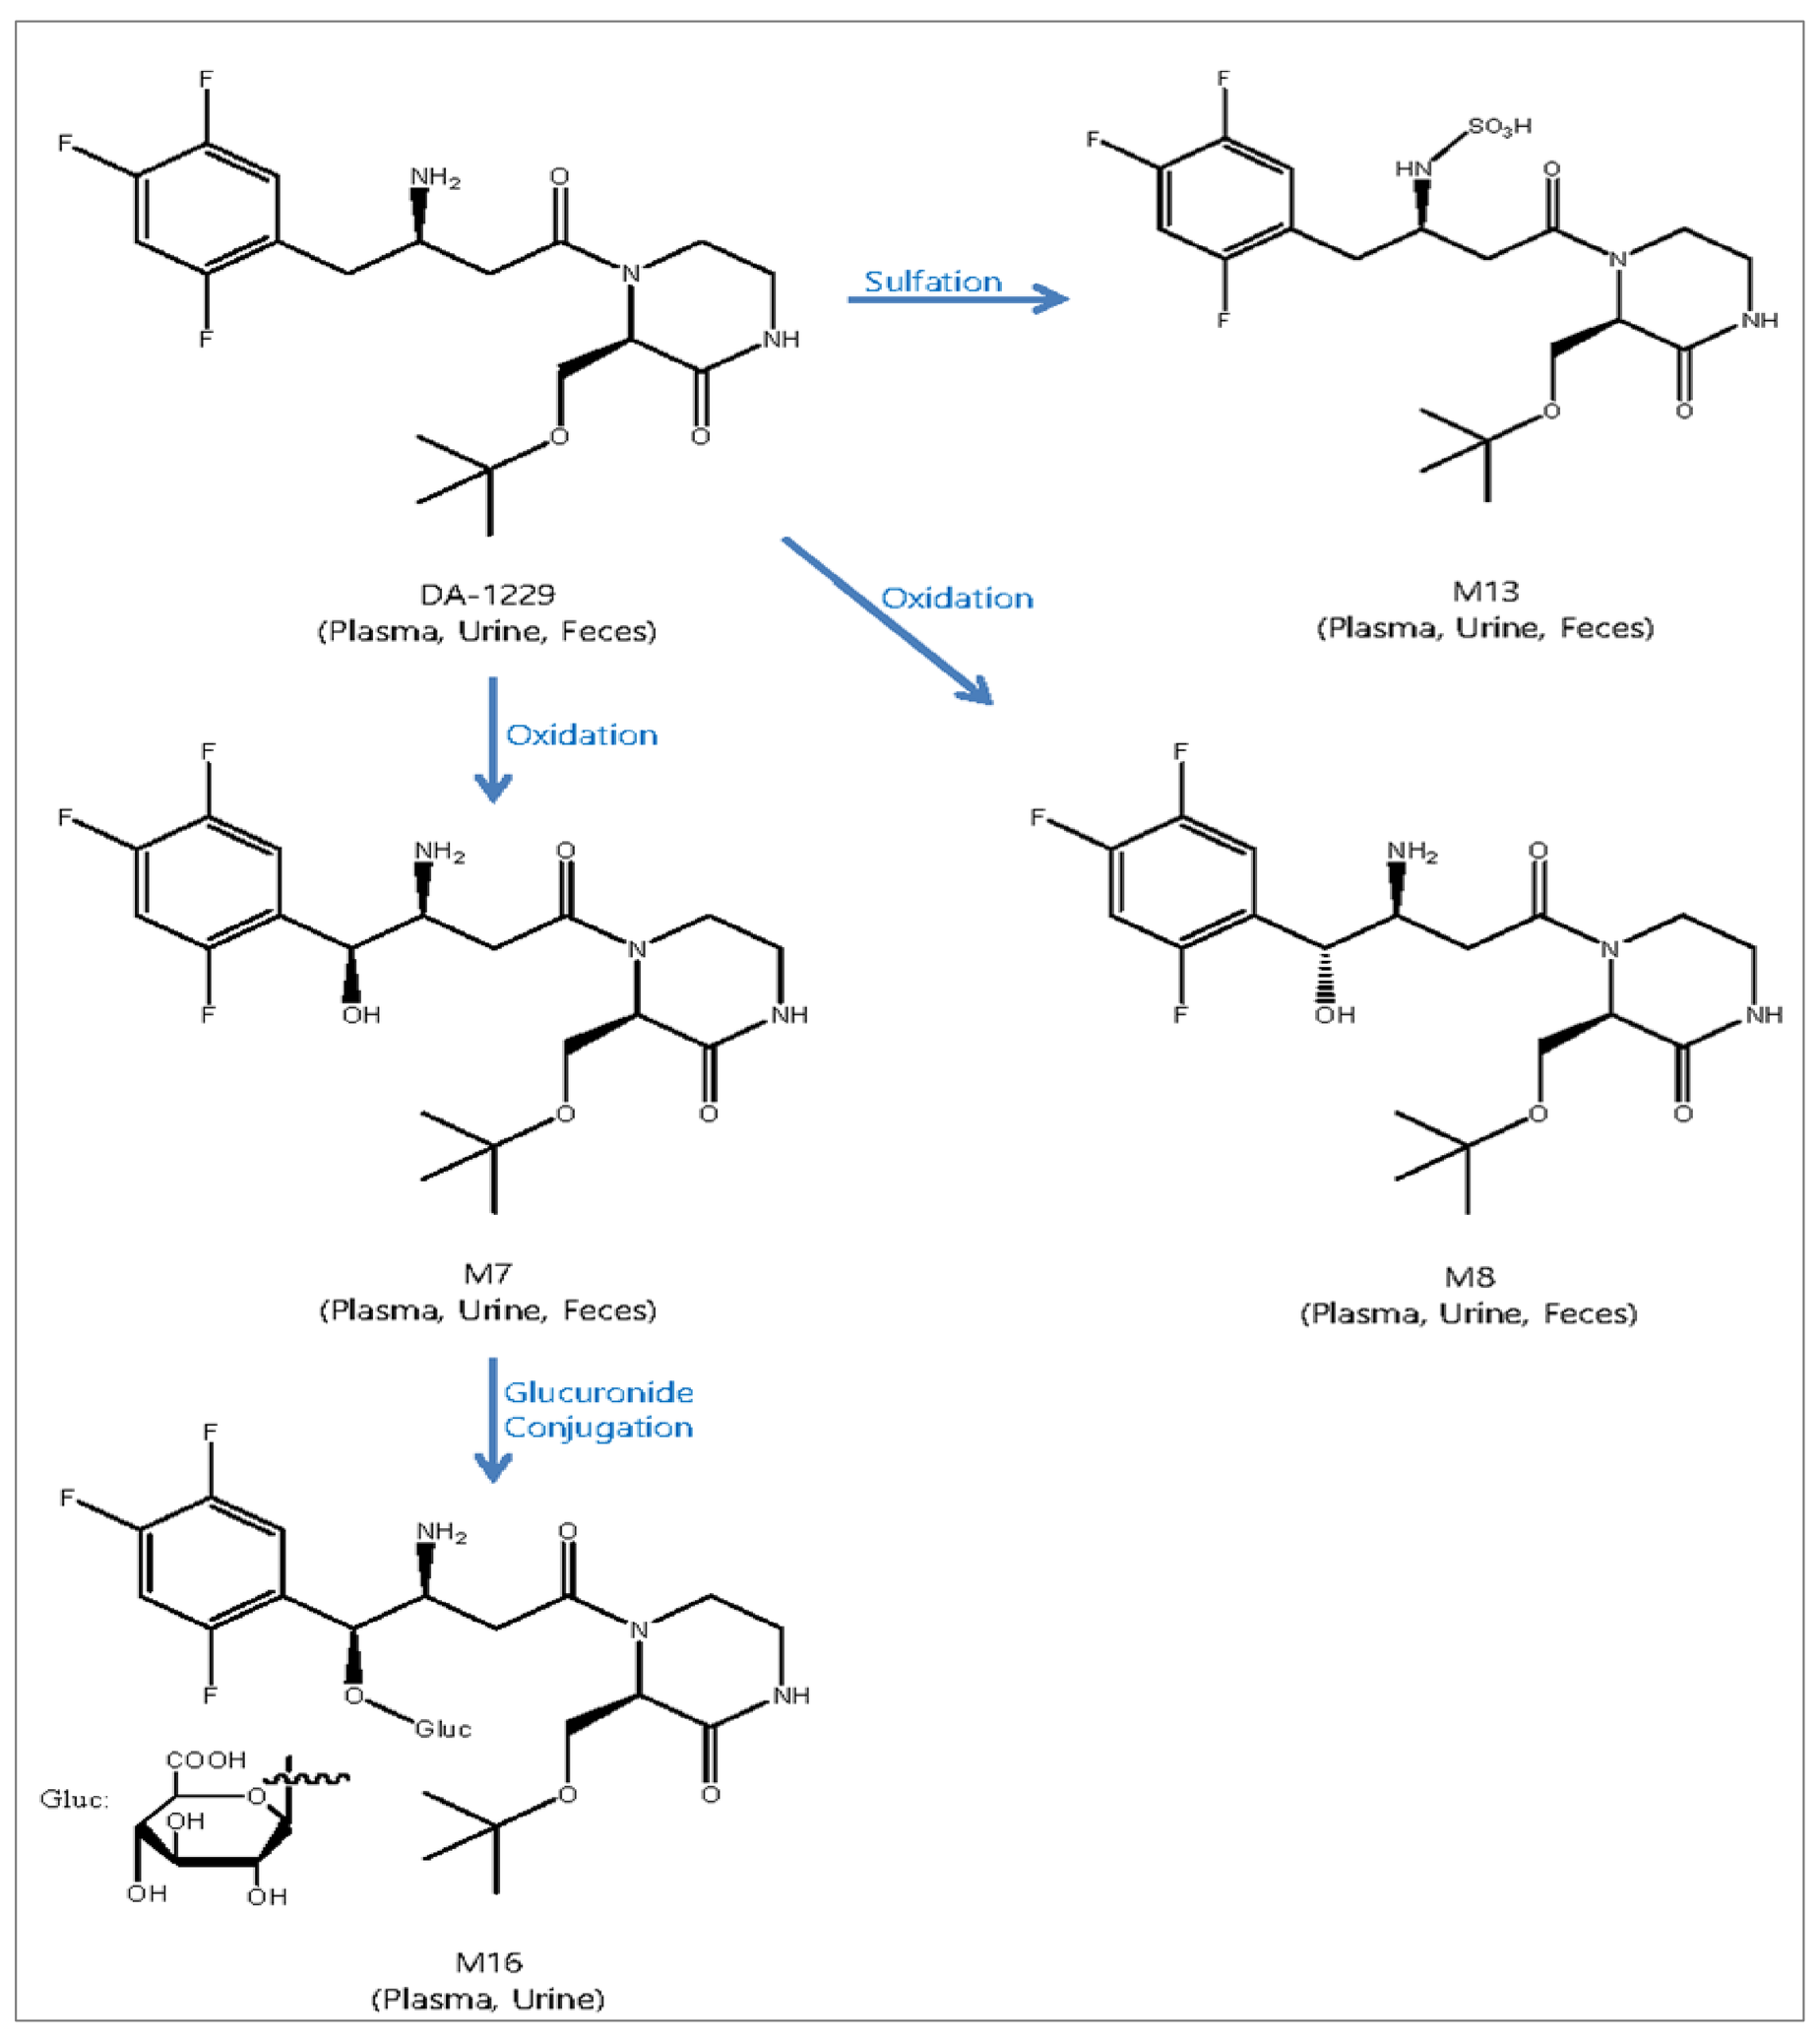
DA-1229
(Plasma, Urine, Feces)
Sulfation
M13
(Plasma, Urine, Feces)
Oxidation
Oxidation
M7
(Plasma, Urine, Feces)
M8
(Plasma, Urine, Feces)
Glucuronide Conjugation
M16
(Plasma, Urine)

Following oral dosing of 5 mg [14C]-Evogliptin the ratio of AUC(0-inf) [14C]-DA-1229:TRA was approximately 52 %. This is a lower percentage than that following IV administration, which suggests pre-systemic metabolism in the form of gut metabolism or first pass loss following oral administration. Mass balance recovery was approximately 89 % over the collection period (10 days). The major circulating component was Evogliptin (71.49%), and no single metabolite accounted for >10% of the circulating TRA. Evogliptin was the major excretion component in urine and faeces, accounting for 56.76% and 57.73% of the TRA, respectively. The major metabolic pathway for Evogliptin was oxidation (M 7 and M8), a sulphated metabolite of Evogliptin was also identified (M13), as was a glucuronide of M7 (M16). Figure 2 presents the proposed metabolic pathways of Evogliptin.

CONCLUSIONS

- Absolute bioavailability of Evogliptin was determined to be approximately 50%.
- Following oral administration of [^{14}C]-Evogliptin, an average of 88.9% of the TRA dose was recovered in excreta; the percentage of TRA excreted via the faeces and urine was similar. Approximately 50 % of the dose was recovered within 48 h in a majority of subjects.
- Evogliptin was the primary contributor to the TRA in plasma, urine and faeces.
- The major metabolic route of Evogliptin was oxidation, with glucuronidation and sulphation also occurring as phase II metabolic routes. Four metabolites were identified.
- This study further demonstrates the value of ^{14}C tracer applications supported by AMS analysis for the evaluation of absolute bioavailability, mass balance and metabolism assessments.

11th International ISSX Meeting

+44 115 9749000 (UK)
+1 800 769 3518 (USA)
www.quotientclinical.com
info@quotientclinical.com